SAN ANTONIO BREAST CANCER SYMPOSIUM
December 10-14, 2019
Henry B. Gonzalez Convention Center, San Antonio, Texas, USA

UNIVERSITEIT VAN PRETORIA
UNIVERSITY OF PRETORIA
YUNIBESITHI YA PRETORIA

The Medical Oncology Centre of Rosebank
Personalised Cancer Care

Tumour Infiltrating Lymphocytes (TILs) in triple-negative breast cancer: High Immunoscore is associated with pathological CR in patients receiving neoadjuvant chemotherapy

BL Rapoport [1,2], S Nayler [3], J Galon [4], B Mlecnik [4], T Smit [1], J Barnard-Tidy [1], A Fugon [4], M Martel [4], R. Anderson [2], CA Benn [5]

[1] The Medical Oncology Centre of Rosebank, Johannnesburg, South Africa; [2] Department of Immunology, Faculty of Health Sciences, University of Pretoria, South Africa; [3] Gritzman and Thatcher Inc. Laboratories, Johannesburg, South Africa; [3] Wits Donald Gordon Medical Centre, Johannesburg, South Africa; [4] HalioDx, Marseille, France; [5] Breast Care Centre, Head of Helen Joseph Hospital Breast Centre Johannnesburg, South Africa

Drs Gritzman & Thatcher Inc. Laboratories
BREAST CARE CENTRE A Specialised Unit
halio dx

Introduction

Background

- Neoadjuvant chemotherapy is widely used to downstage breast cancers prior to surgery.
- Pathologic complete response (pCR) rate is a strong predictor of outcome for breast cancer.

Immunoscore®

- The Immunoscore® assay is the first standardized immune-based assay for classification of cancer [Hermitte et al., 2016]. It assesses the host immune response by measuring intra- and peri-tumoral T cell infiltration in formalin-fixed paraffin-embedded (FFPE) tissue sections.
- Originally developed for colon cancer indication, it is intended to be widely used in solid cancer indications for diagnostic and prognostic purposes, as well as a pharmacodynamic biomarker during drug development processes. As a first clinical validation in breast cancer, we assessed the Immunoscore in a cohort of 103 breast cancer patients, that previously received neo-adjuvant chemotherapy.

Methods

Pathological and clinical assessment

- Clinical assessment of the primary tumour and lymph nodes was made using bi-dimensional caliper measurements of the primary tumour and axillary nodes.
- Sonographical assessments of the primary tumour and lymph nodes were performed regularly.
- Immunohistochemical staining was performed for ER, PR, HER-2 and Ki67 .
- Fluorescence in situ hybridization (FISH) was used to confirm HER-2 positivity.
- We analyzed data retrospectively/prospectively on 103 breast cancer patients undergoing neoadjuvant chemotherapy.
- Pathological complete response (pCR) was defined as the complete disappearance of the invasive cancer in the breast and absence of tumour in the axillary lymph nodes.
- Ethics approval was obtained from Pharma-Ethics, Pretoria, South Africa (ethics committee working according to the South African Ethics regulations).
- NCSS software version 11 for Windows (USA) was used for statistical analyses.
- Outcome assessments: Associations of clinical and pathological characteristics including Ki67, CD8+ cytotoxic T cells and CD3+ T cells with pCR.
- All patients were treated with anthracycline and/or taxane-based neoadjuvant chemotherapy.

Immunoscore® Assessment

- In this retrospective analysis, 103 pre-treatment tumour tissue samples were analyzed by immunohistochemistry for density (cells/mm³) of T-cell subsets (CD3+,CD8+).
- CD3 and CD8 staining was performed using Benchmark® XT station on 2 consecutive formalin-fixed paraffin-embedded (FFPE) slides (4 µm).
- Digital pathology-dedicated software permitted the measurement of positive cell densities into interest area (core of the tumour and invasive margin).
- A prespecified bioinformatics algorithm was used to generate a numerical index (Immunoscore®) and analysis cut-offs. Immunoscore® assay measures the density of CD8+ cytotoxic T cells and CD3+ T cells of resected or biopsied cancer samples and performed on FFPE tissue slides.
- Immunoscore® provides 3 score levels (high / intermediary / low).
- Immunoscore® was applied to tumours with invasive margin and was adapted when no invasion was identified on the specimen.

Figure 1. Immunoscore® Assessment.

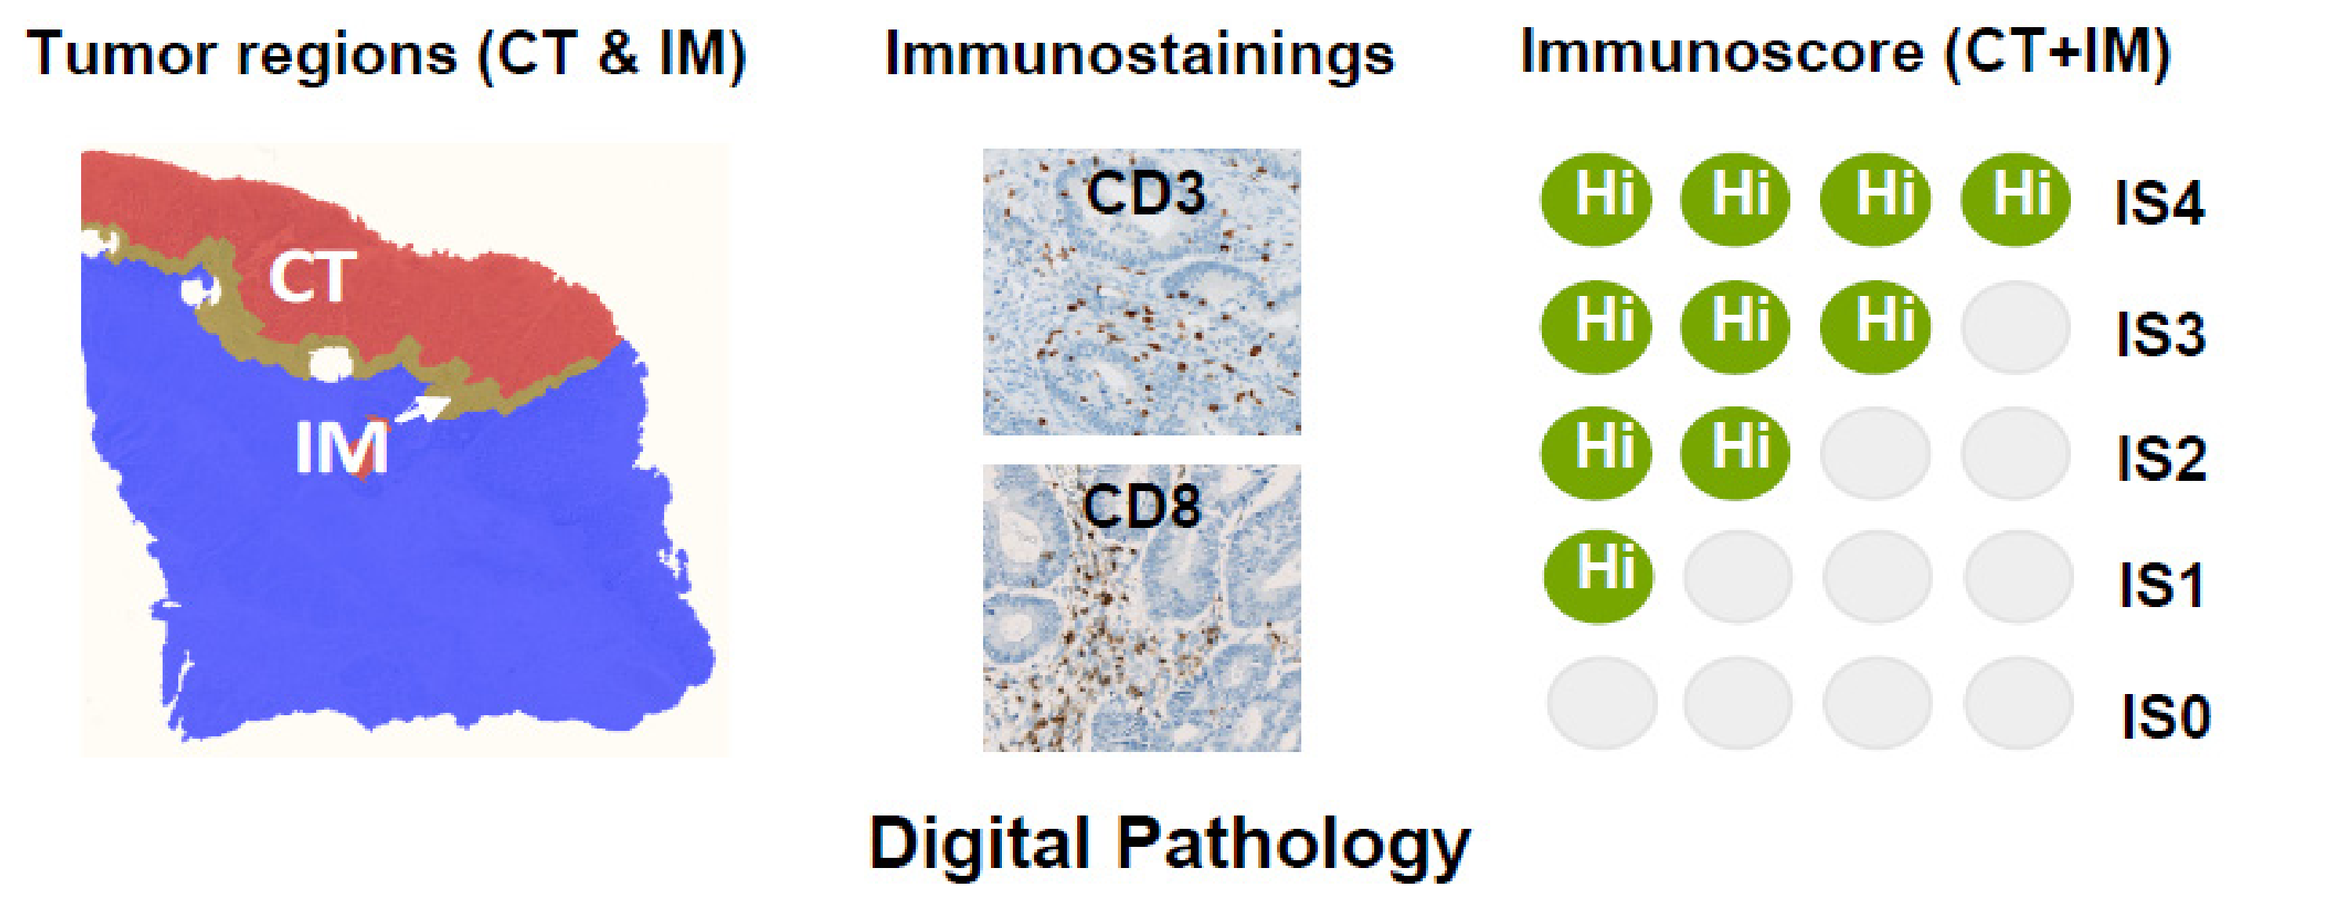
Tumor regions (CT & IM)
CT
IM
Immunostainings
CD3
CD8
Immunoscore (CT+IM)
Hi Hi Hi Hi IS4
Hi Hi Hi IS3
Hi Hi IS2
Hi IS1
IS0
Digital Pathology

Figure 2. Immunoscore® High.

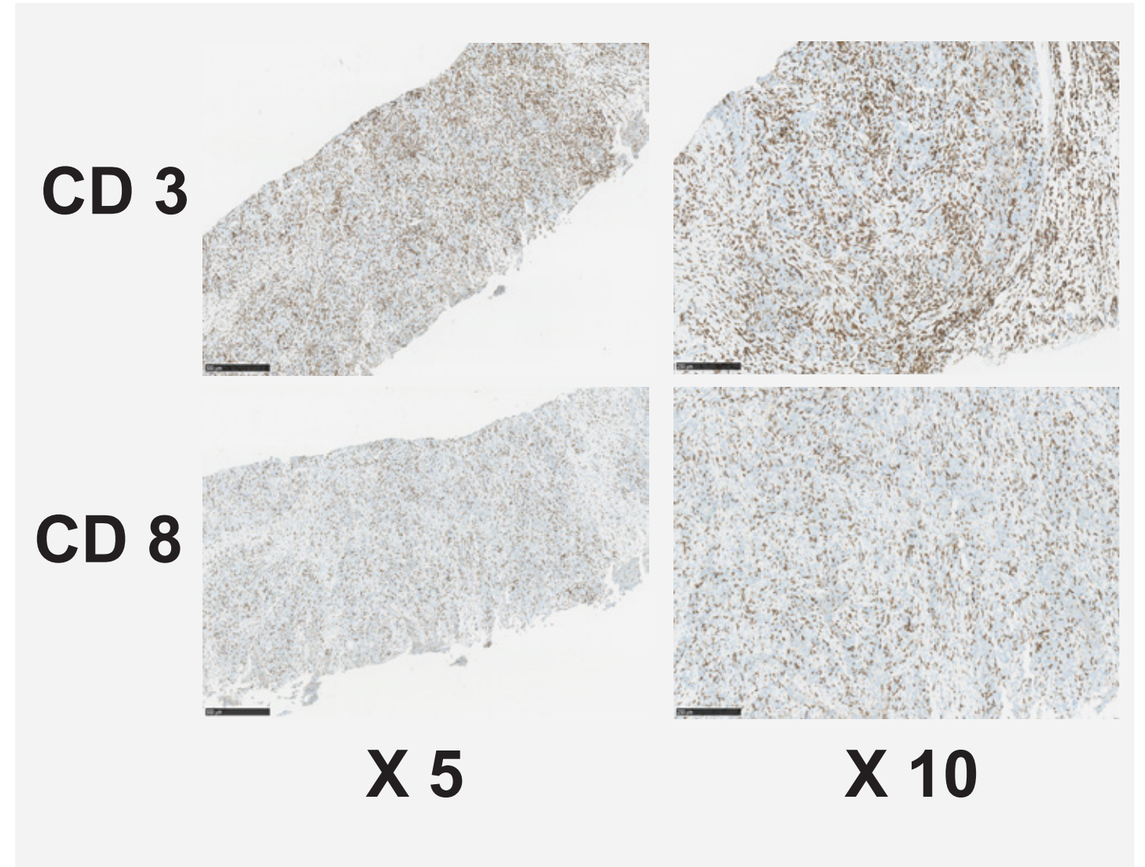
CD 3
CD 8
X 5
X 10

Figure 3. Immunoscore® Low.

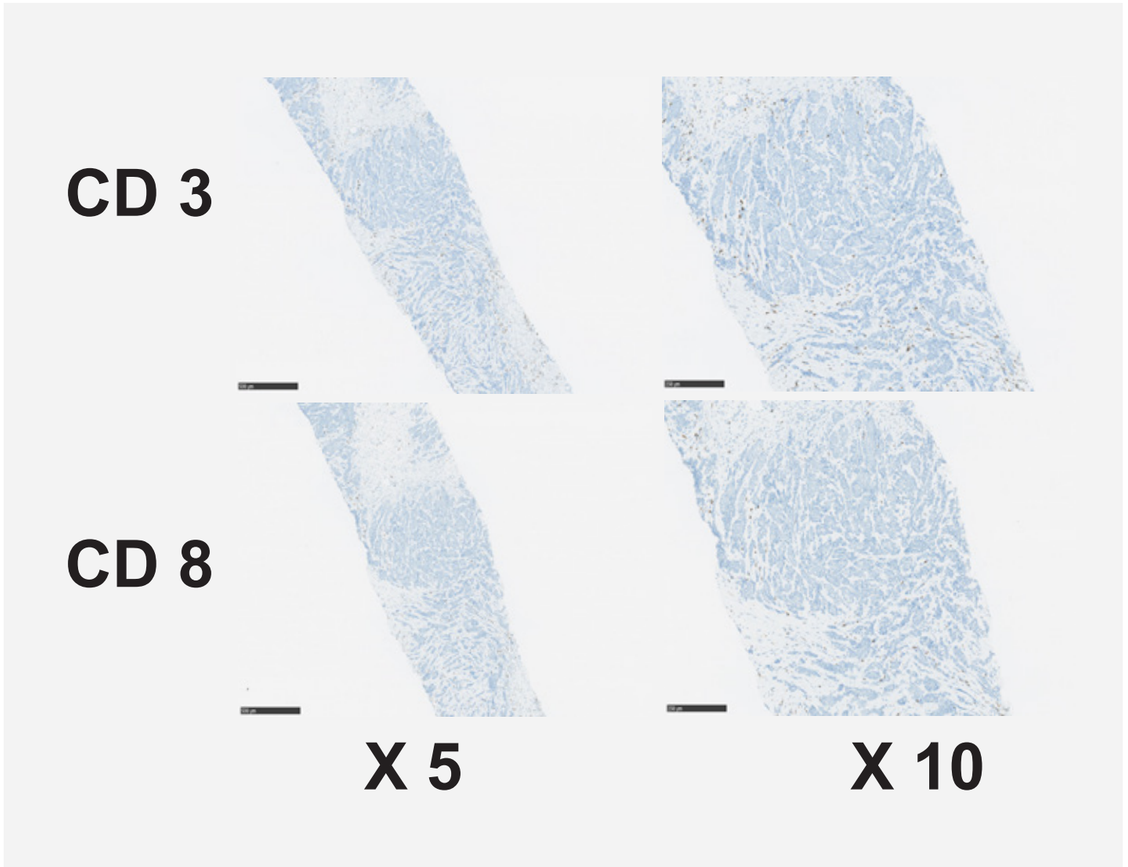
CD 3
CD 8
X 5
X 10

Figure 4. Invasive margin.

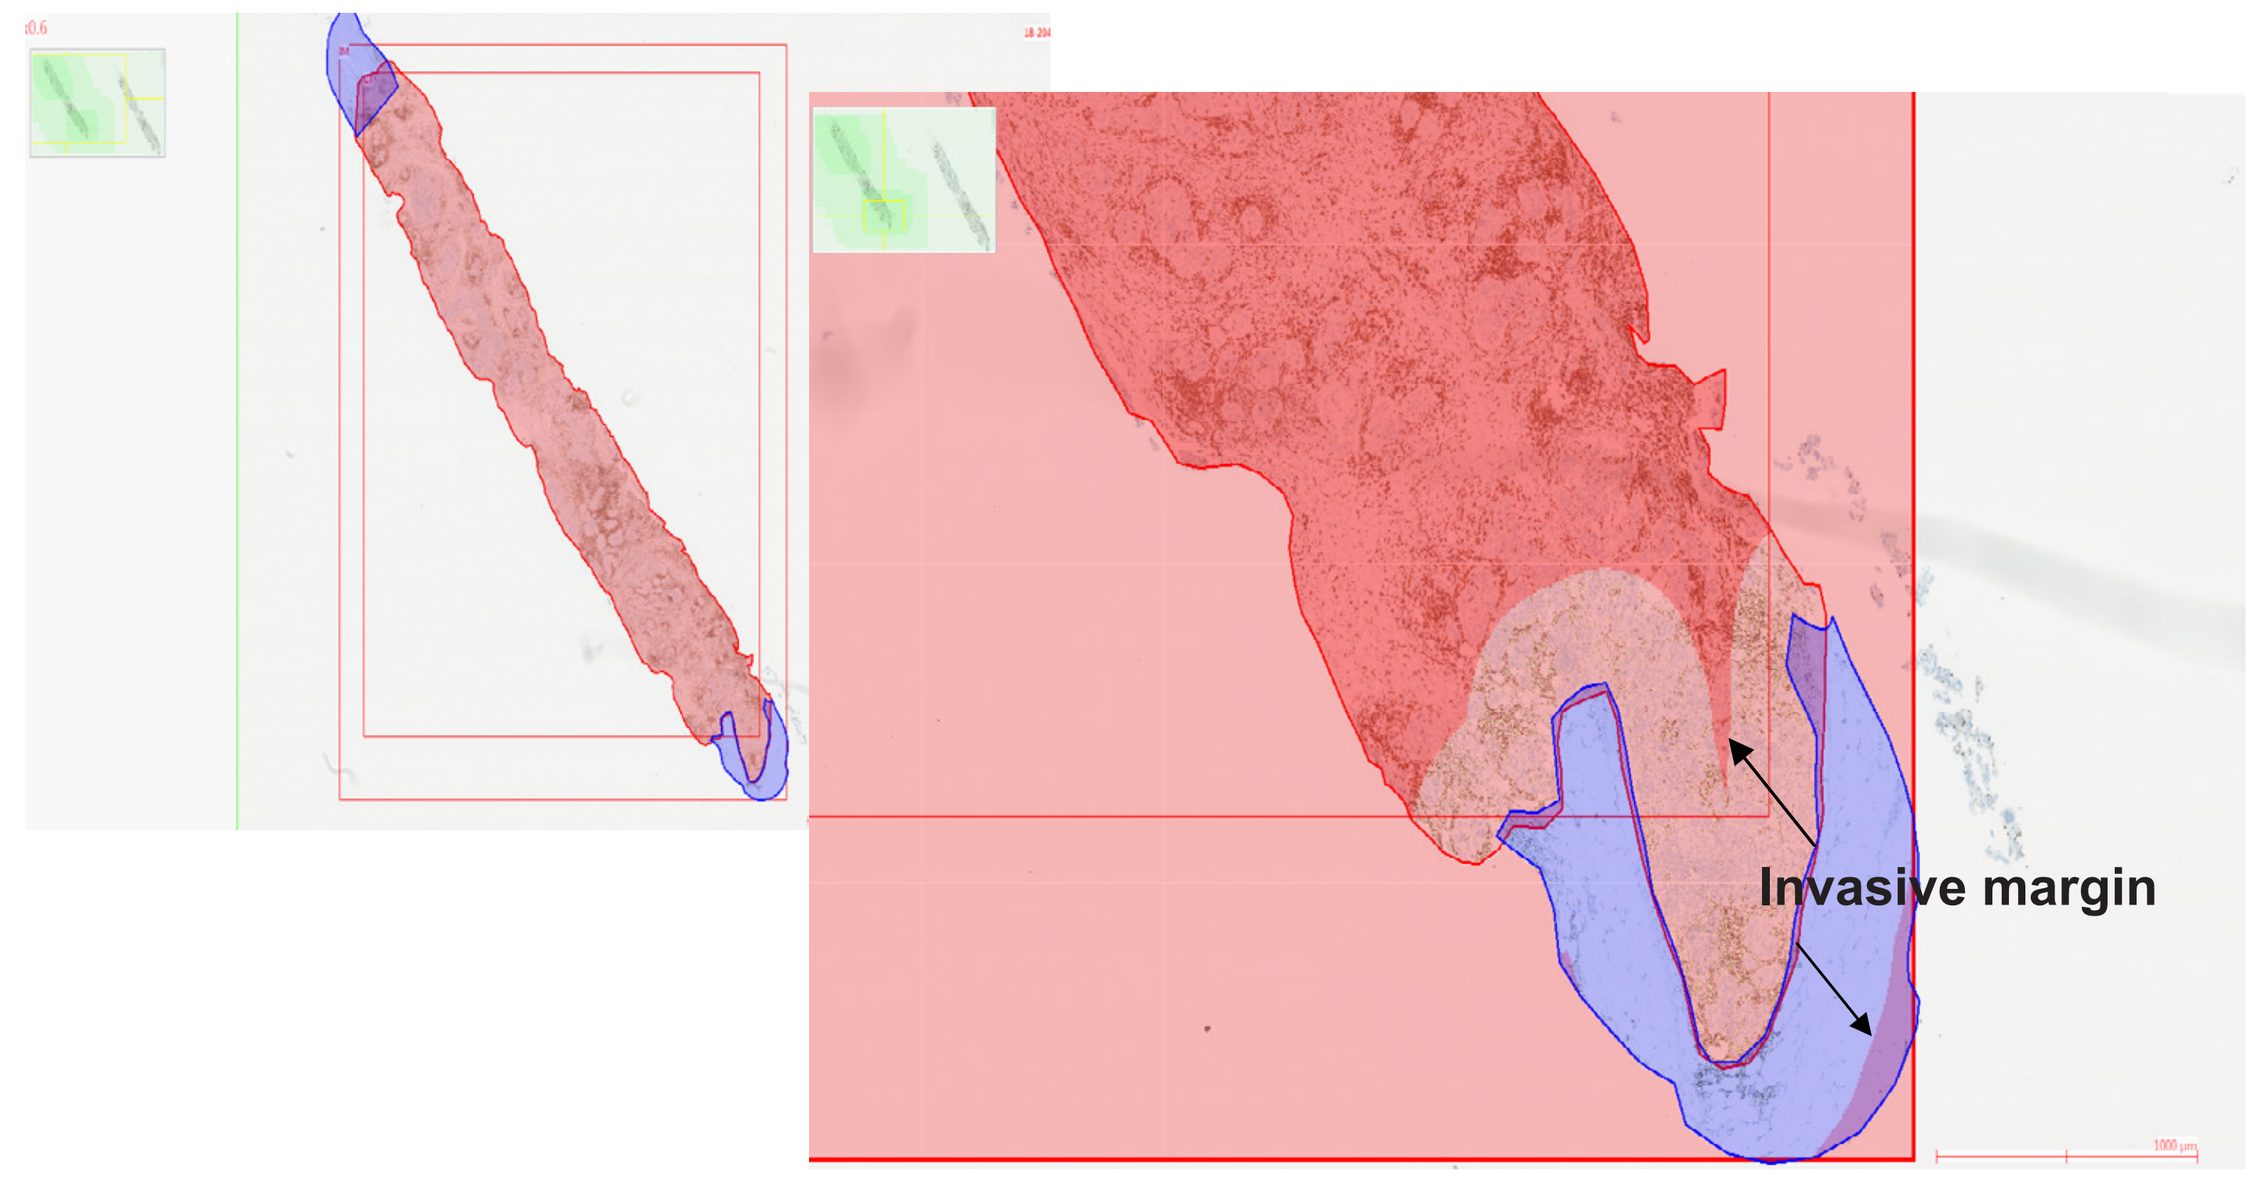
Invasive margin

Statistical Methods

- The primary hypothesis was that higher levels of CD8+ cytotoxic T cells, CD3+ T cells and Immunoscore® would be associated with a better overall prognosis, independent of anti-cancer therapy.
- The Mann Whitney U-test was used to compare the cell density between TNBC and Non-TNBC patients.
- Receiver-operating characteristic (ROC) curve analysis was used to determine the optimal cut-point for Ki67, CD8+ cytotoxic T cells, CD3+ T cells and Immunoscore®.
- Fisher's exact or Chi-squared tests were used for the analysis of categorical variables.
- Logistic regression multivariate models included only variables that exhibited a univariate association with the dependent variable, pCR (p < 0.1).
- NCSS software version 11 for Windows (USA) was used for statistical analyses.

Patient Characteristics

Table 1. Patient Characteristics.

Age (n=103)		
Median Age	52	
Range	26-84	
Histology		
	Total	%
Ductal	99	96%
Lobular	2	2%
Other	2	2%
Menepausal Status		
	Total	%
Pre	41	40%
Post	62	60%
Tumour Size		
	Total	%
T1	23	22%
T2	65	63%
T3 + T4	15	15%
Nodal Status		
	Total	%
Negative	45	44%
Positive	54	52%
Unknown	4	4%
Stage		
	Total	%
A1	8	7%
A2	1	1%
B1	49	48%
B2	26	25%
C1	10	10%
C2	7	7%
C3	2	2%
Ki-67 Pre-chemo		
Median	40	
Range	5-90	
	Total	%
≥ 40%	51	50%
15-39%	37	35%
< 15%	13	13%
Unknown	2	2%
Molecular type		
	Total	%
Luminal A	9	9%
Luminal B	23	22%
HER2 Positive	18	18%
TNBC	53	51%
CD3 and CD8 Count		
	Median Cells/ mm³	Range Cells/ mm³
CD3 centre of tumour	884	25-5771
CD3 invasive margin	1409	53-6197
CD8 centre of tumour	358	10-3448
CD8 invasive margin	535	38-3117
Immunoscore		
	Total	%
0	8	8%
1	9	8%
2	48	47%
3	35	34%
4	3	3%

Results

ROC Curves for Prediction of pCR

Figure 5. ROC Curve – CD3-Centre of Tumour.

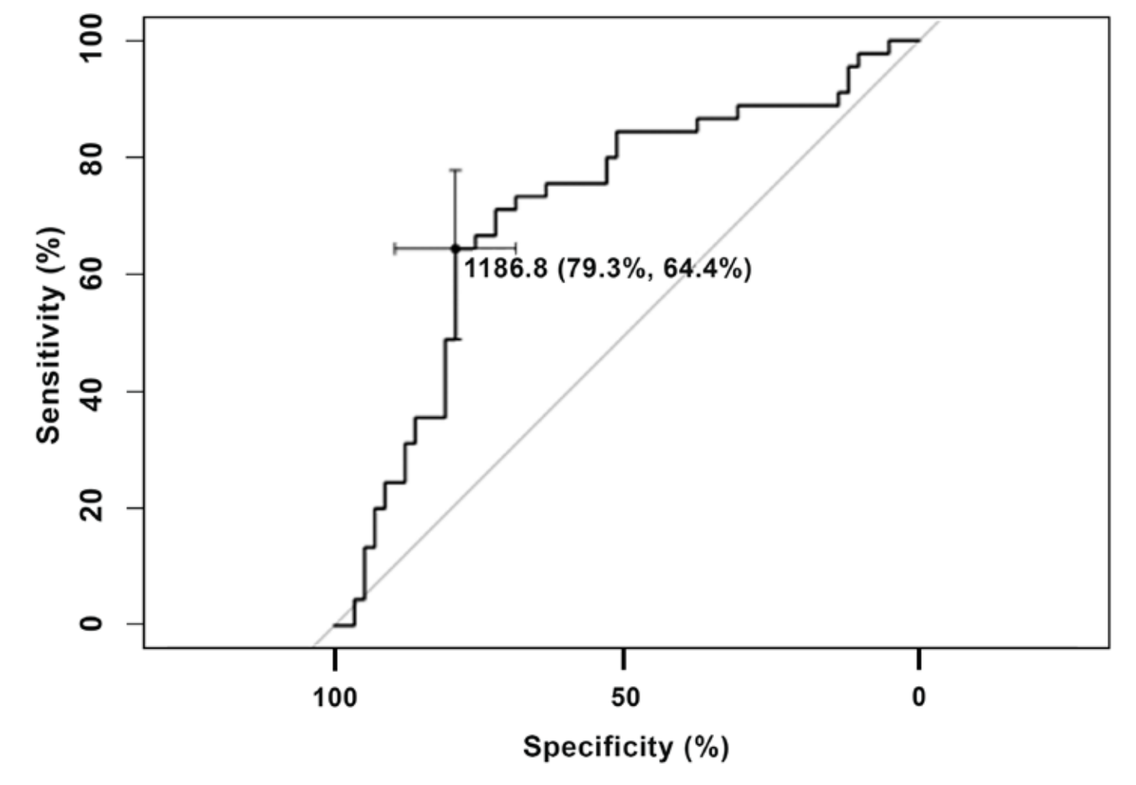
1186.8 (79.3%, 64.4%)
Sensitivity (%)
Specificity (%)

Figure 6. ROC Curve – CD8 – Centre of Tumour.

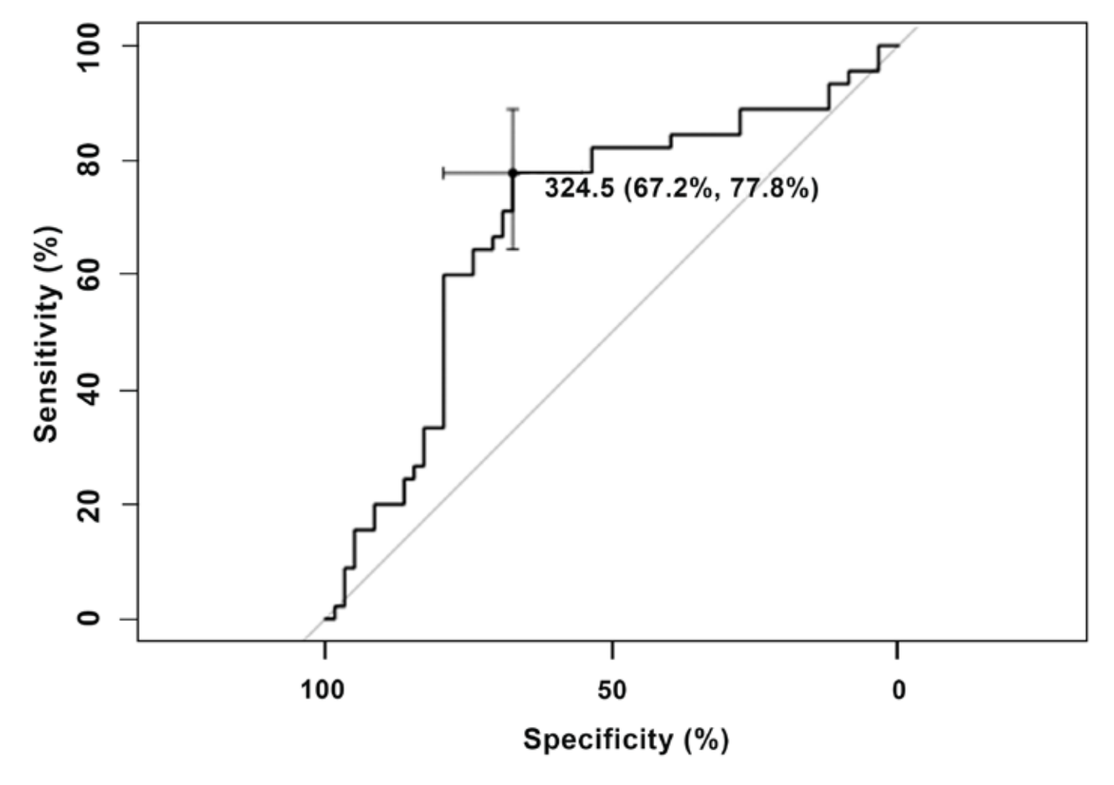
324.5 (67.2%, 77.8%)
Sensitivity (%)
Specificity (%)

Figure 7. ROC Curve – CD3 – Invasive Margin.

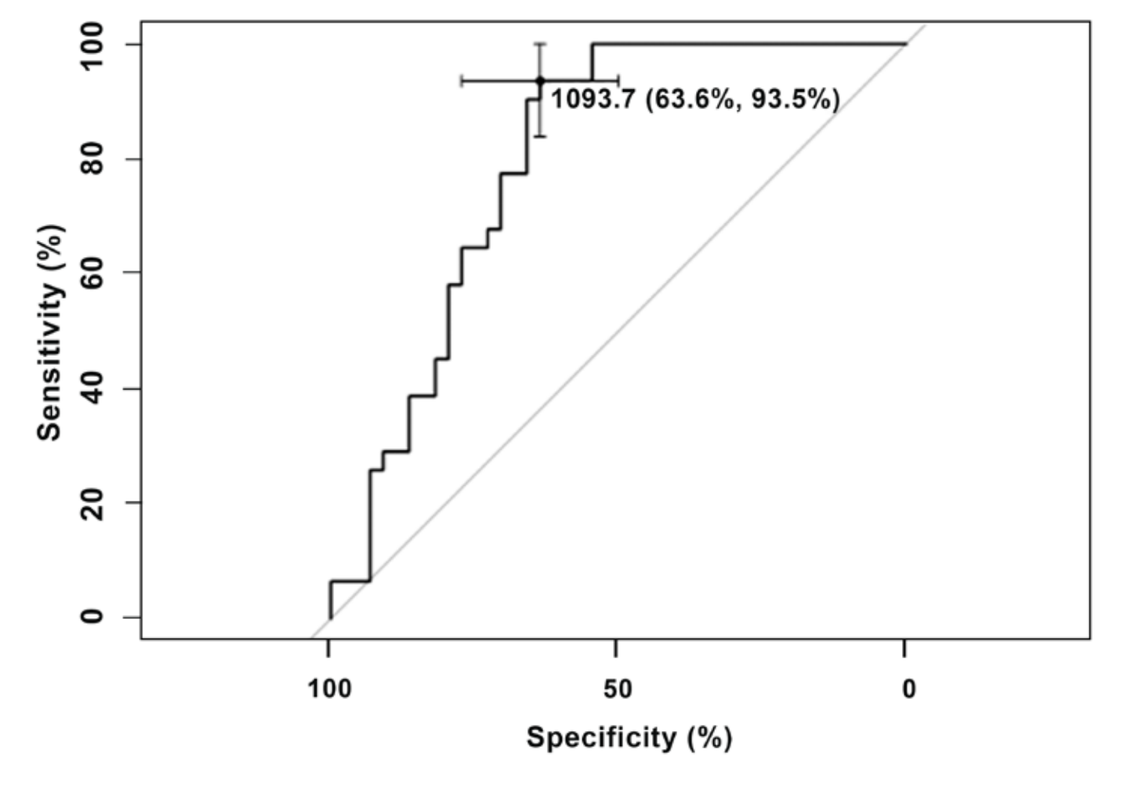
1093.7 (63.6%, 93.5%)
Sensitivity (%)
Specificity (%)

Figure 8. ROC Curve – CD8 – Invasive Margin.

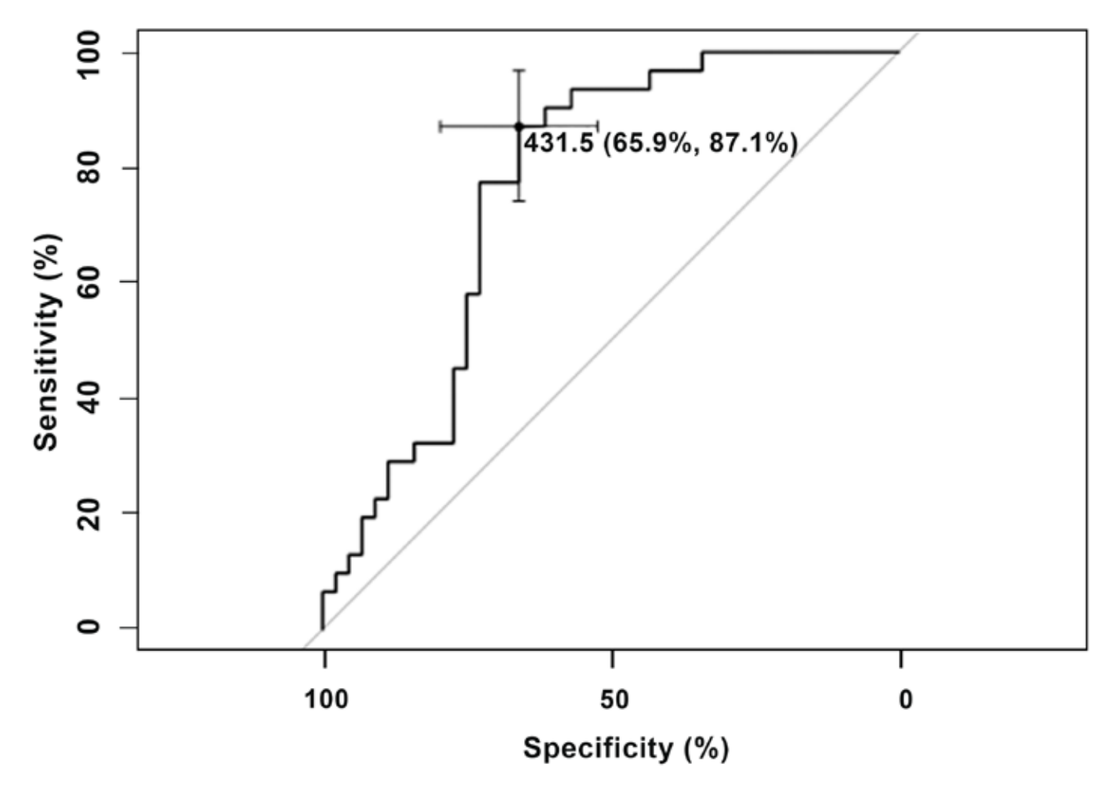
431.5 (65.9%, 87.1%)
Sensitivity (%)
Specificity (%)

Figure 9. Percentage of patients with cell density below/over 1200 mm³ (Centre of Tumour).

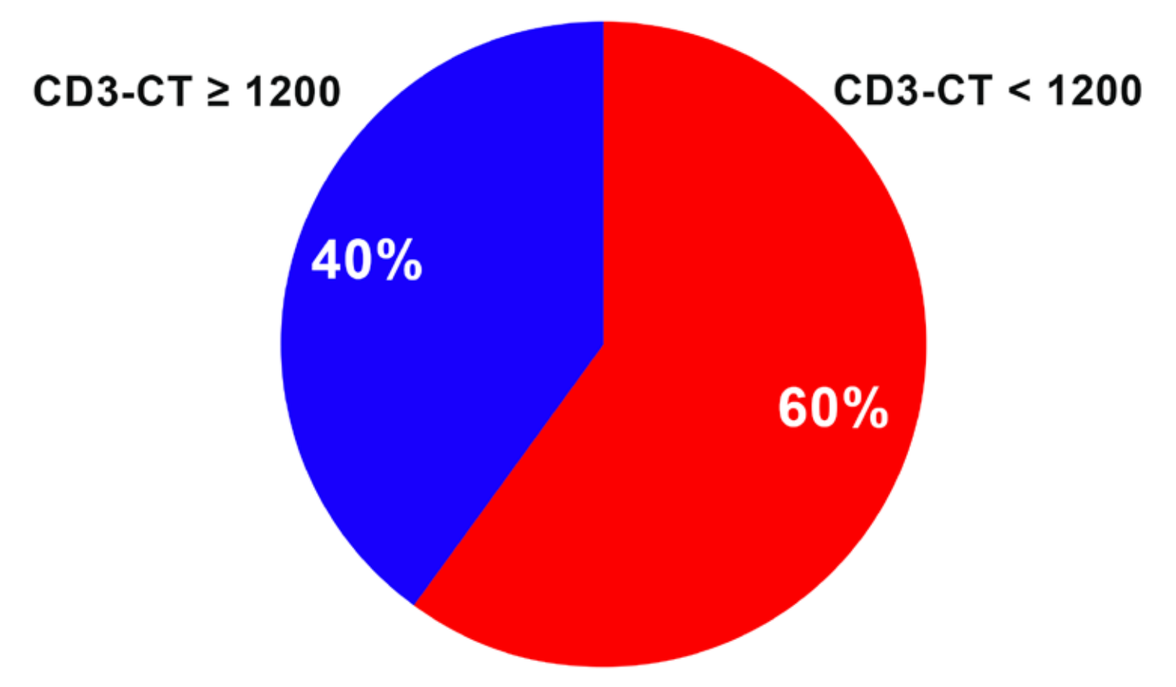
CD3-CT ≥ 1200
CD3-CT < 1200
40%
60%

Figure 10. Percentage of patients with cell density below/over 1100 mm³ (Invasive Margin).

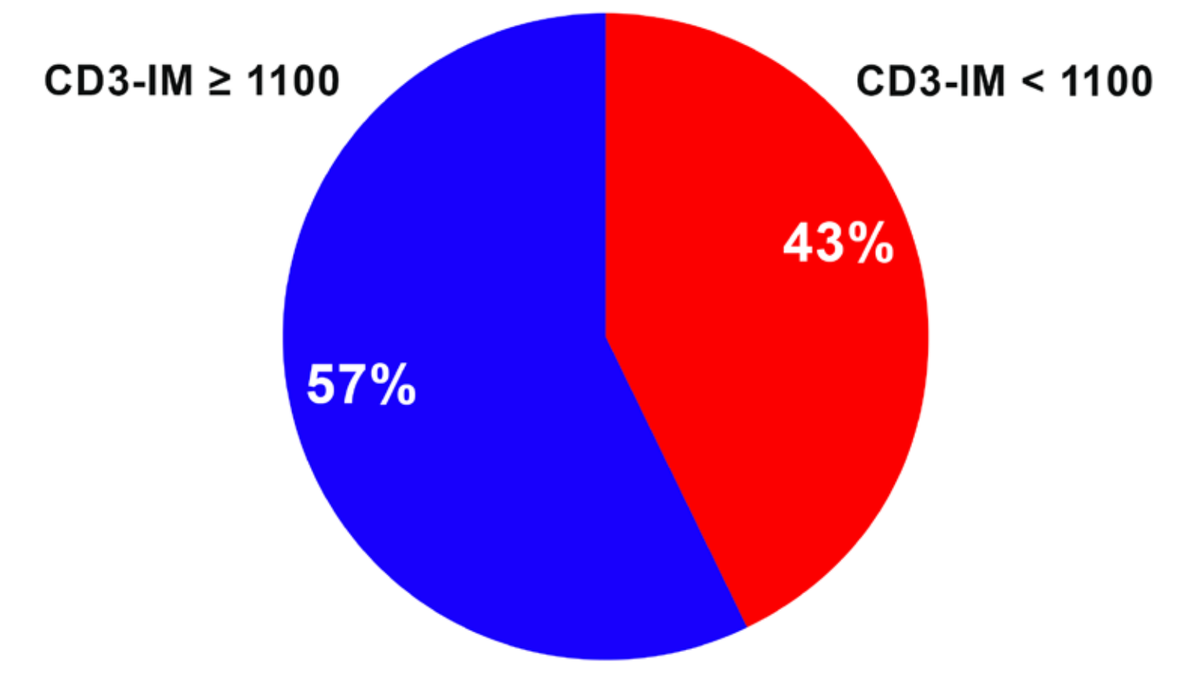
CD3-IM ≥ 1100
CD3-IM < 1100
57%
43%

Figure 11. Percentage of patients with cell density below/over 300 mm³ (Centre of Tumour).

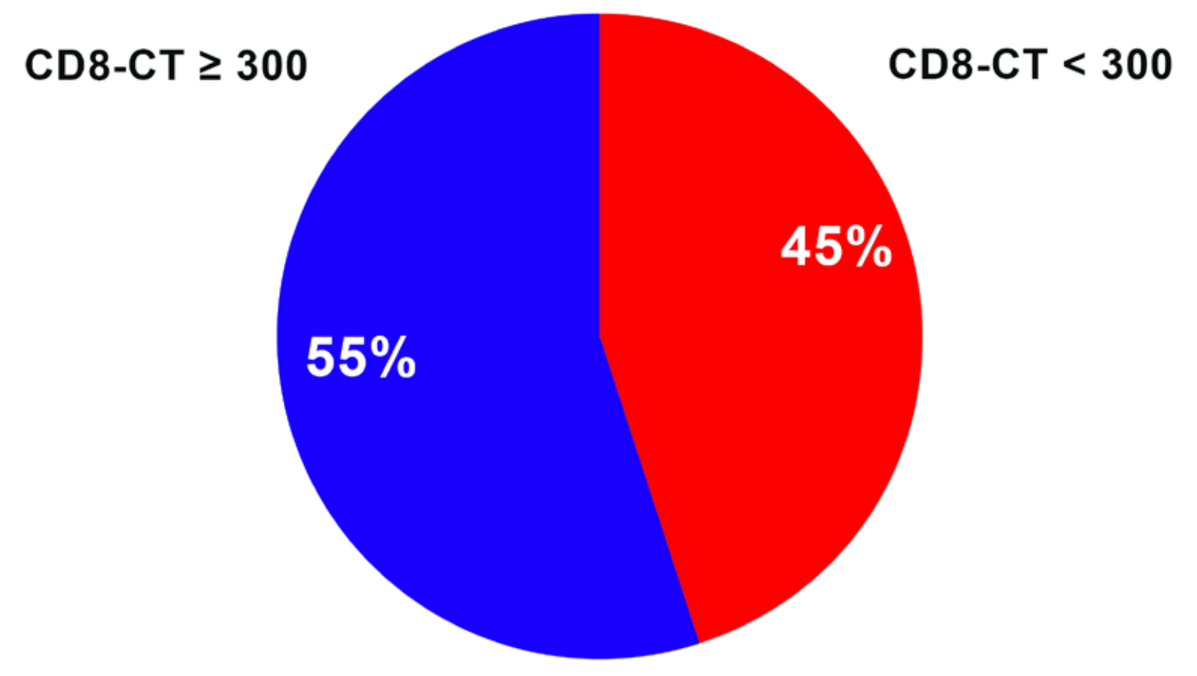
CD8-CT ≥ 300
CD8-CT < 300
55%
45%

Figure 12. Percentage of patients with cell density below/over 300 mm³ (Centre of Tumour).

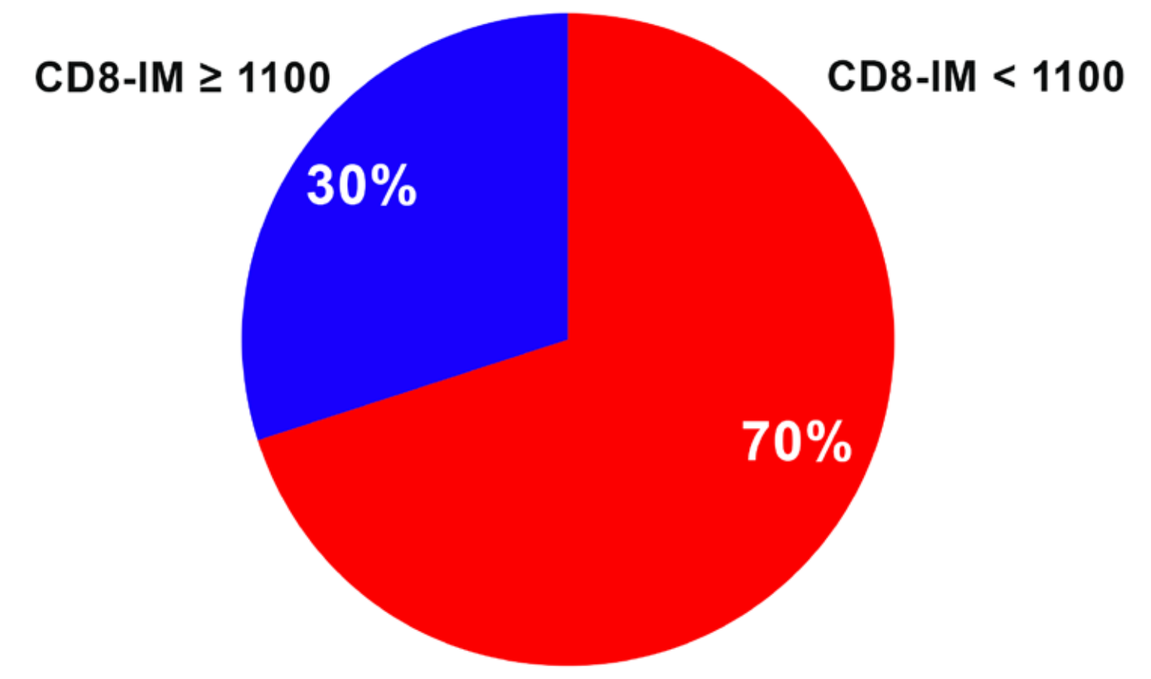
CD8-IM ≥ 1100
CD8-IM < 1100
30%
70%

T-Cell densities compare between TNBC vs Non-TNBC patients

Figure 13a. CD3 - Centre of Tumour.

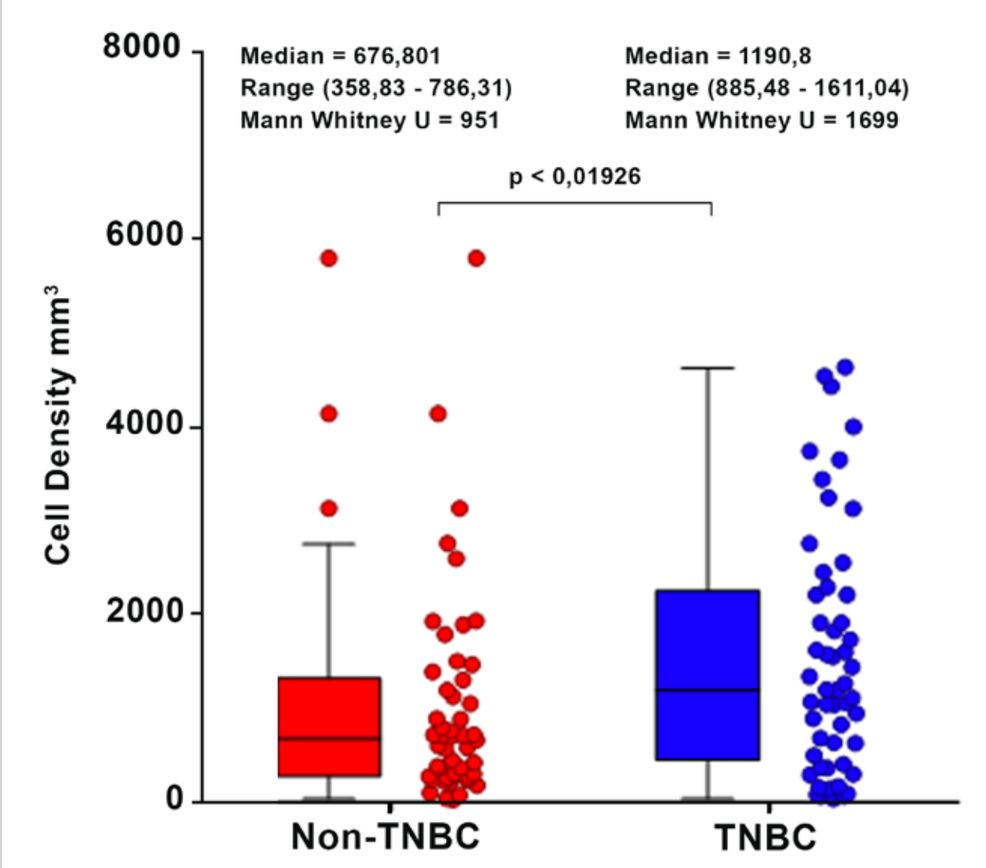
Median = 676,801
Range (358,83 - 786,31)
Mann Whitney U = 951
Median = 1190,8
Range (885,48 - 1611,04)
Mann Whitney U = 1699
p < 0,01926
Cell Density mm³
Non-TNBC
TNBC

Figure 13b. CD3 - Invasive Margin.

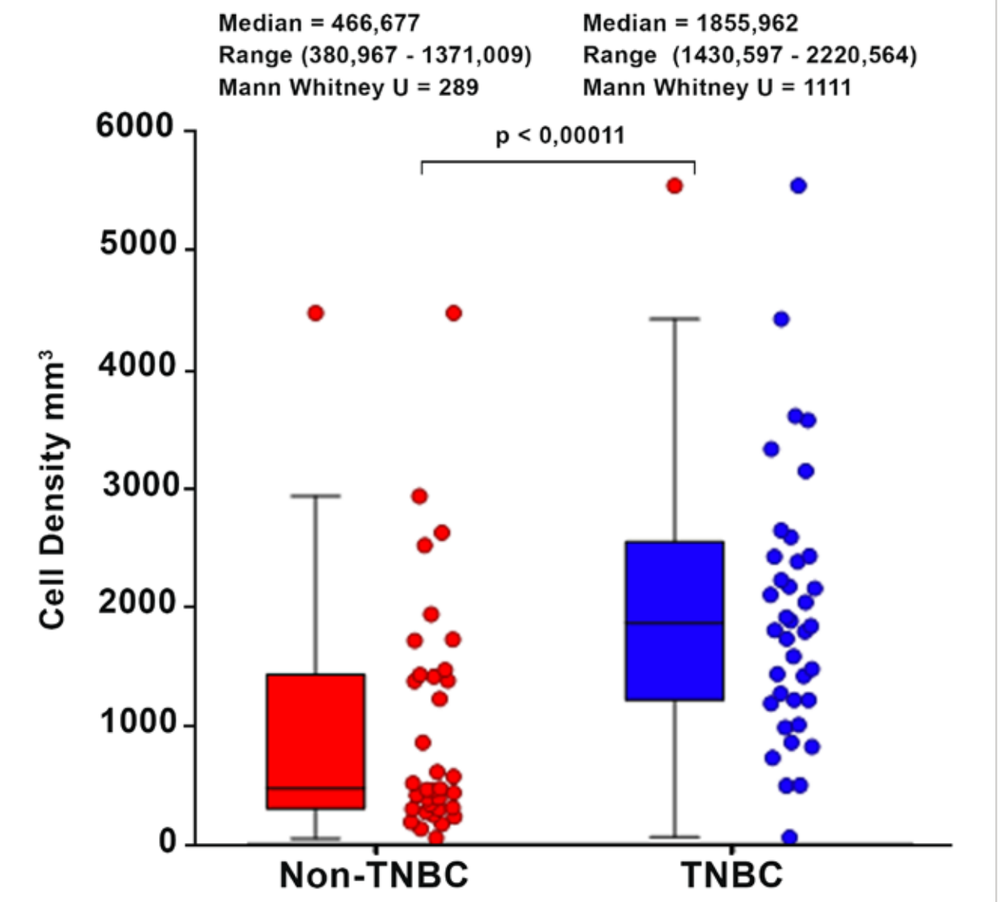
Median = 466,677
Range (380,967 - 1371,009)
Mann Whitney U = 289
Median = 1855,962
Range (1430,597 - 2220,564)
Mann Whitney U = 1111
p < 0,00011
Cell Density mm³
Non-TNBC
TNBC

Figure 13c. CD8 - Centre of Tumour.

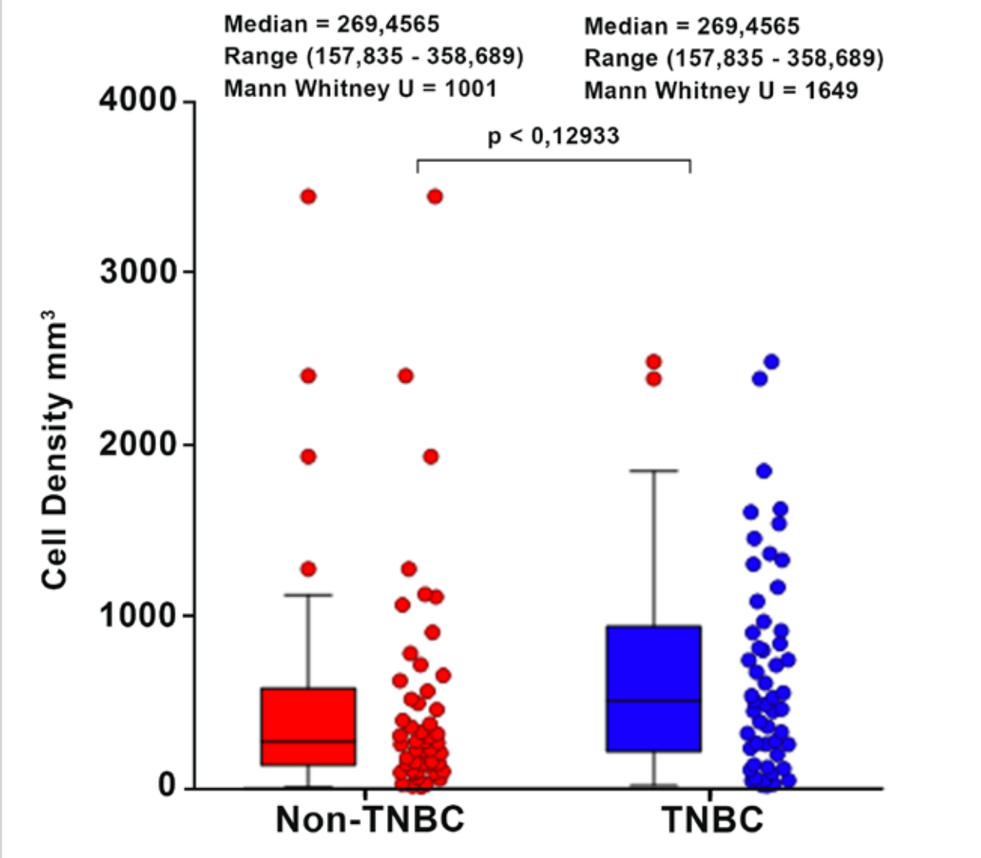
Median = 269,4565
Range (157,835 - 358,689)
Mann Whitney U = 1001
Median = 269,4565
Range (157,835 - 358,689)
Mann Whitney U = 1649
p < 0,12933
Cell Density mm³
Non-TNBC
TNBC

Figure 13d. CD8 - Invasive Margin.

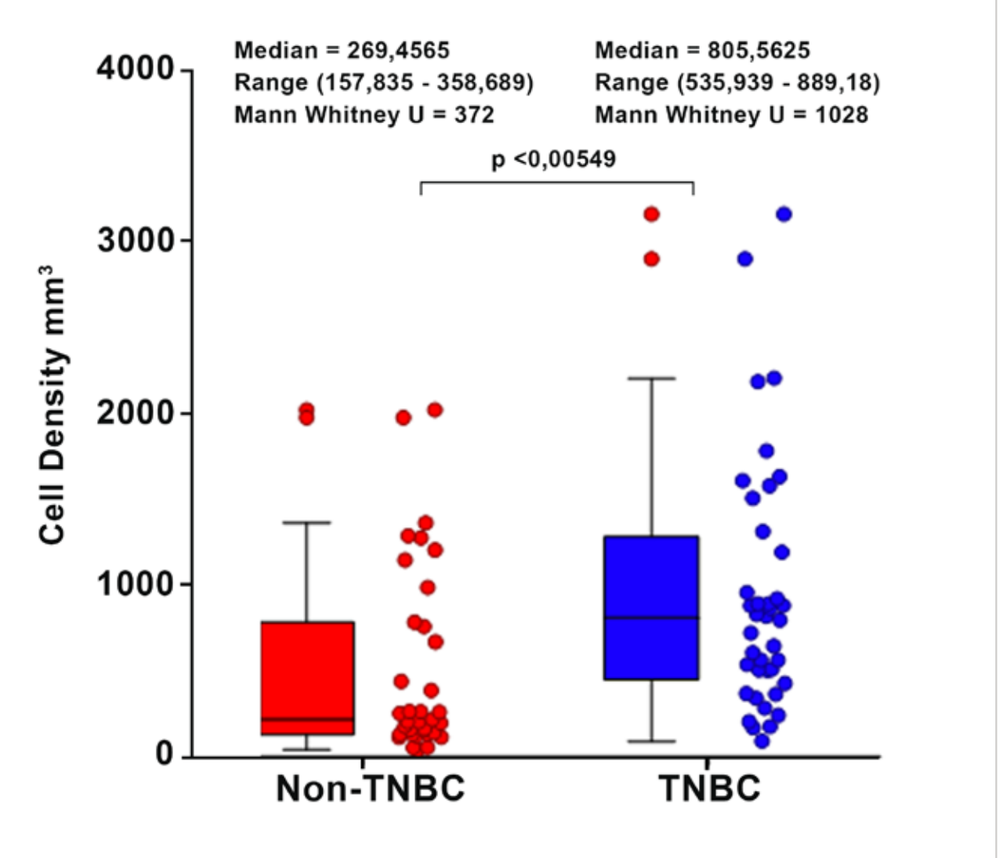
Median = 269,4565
Range (157,835 - 358,689)
Mann Whitney U = 372
Median = 805,5625
Range (535,939 - 889,18)
Mann Whitney U = 1028
p <0,00549
Cell Density mm³
Non-TNBC
TNBC

Univariate Analysis

Table 2. Univariate Analysis - Significant factors assosiated with pCR.

	pCR	Chi²	pValue
Stage			
1	67%	9,03	0.02885
2A	51%		
2B	42%		
3	16%		
ER			
Positive	18%	21.80	0.00001
Negative	64%		
PR			
Positive	13%	22.81	0.00001
Negative	61%		
HER2			
Postive	67%	0.3531	0.55237
Negative	51%		
Molecular type			
Luminal	9%	23,03	0,00001
HER2 Positive	50%		
TNBC	62%		
Ki-67			
≥ 40%	57%	13,84	0,00099
15-39%	41%		
< 15%	0%		
Immunoscore®			
High	63%	9,99	0,00674
Intermediate	35%		
Low	23%		
Immunoscore®			
High	63%	9,27	0,00010
Intermediate + Low	32%		

Figure 14. Response to Neo-Adjuvant Chemotherapy.

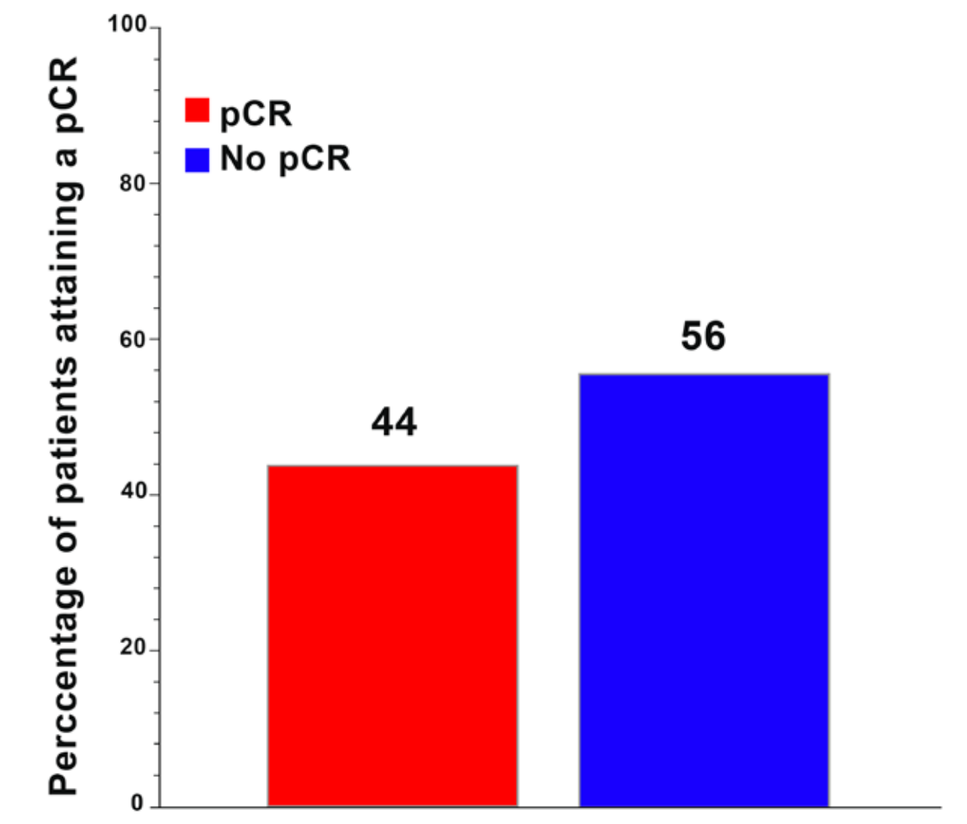
pCR
No pCR
44
56
Perccentage of patients attaining a pCR

Figure 15. Percentage of patients attaining a pCR.

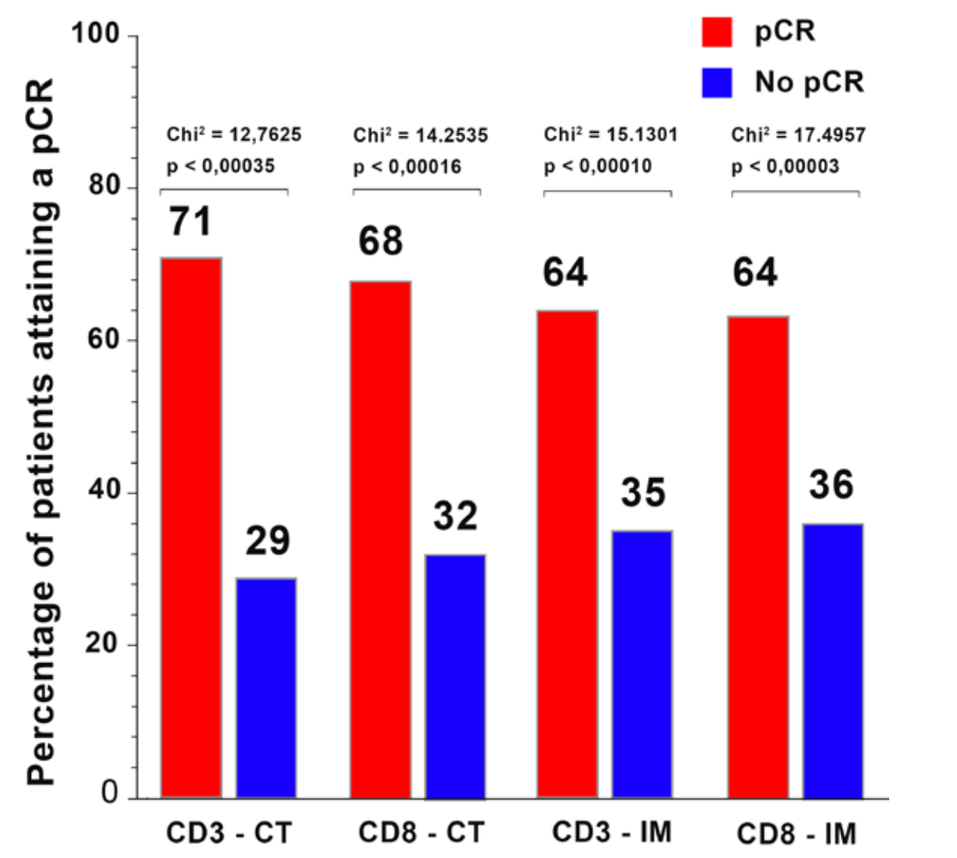
pCR
No pCR
Chi² = 12,7625 p < 0,00035
Chi² = 14.2535 p < 0,00016
Chi² = 15.1301 p < 0,00010
Chi² = 17.4957 p < 0,00003
71
29
68
32
64
35
64
36
Percentage of patients attaining a pCR
CD3 - CT
CD8 - CT
CD3 - IM
CD8 - IM

Median cell density in patients with pCR vs non-pCR patients

Figure 16. Median cell density in patients with pCR vs non-pCR patients.

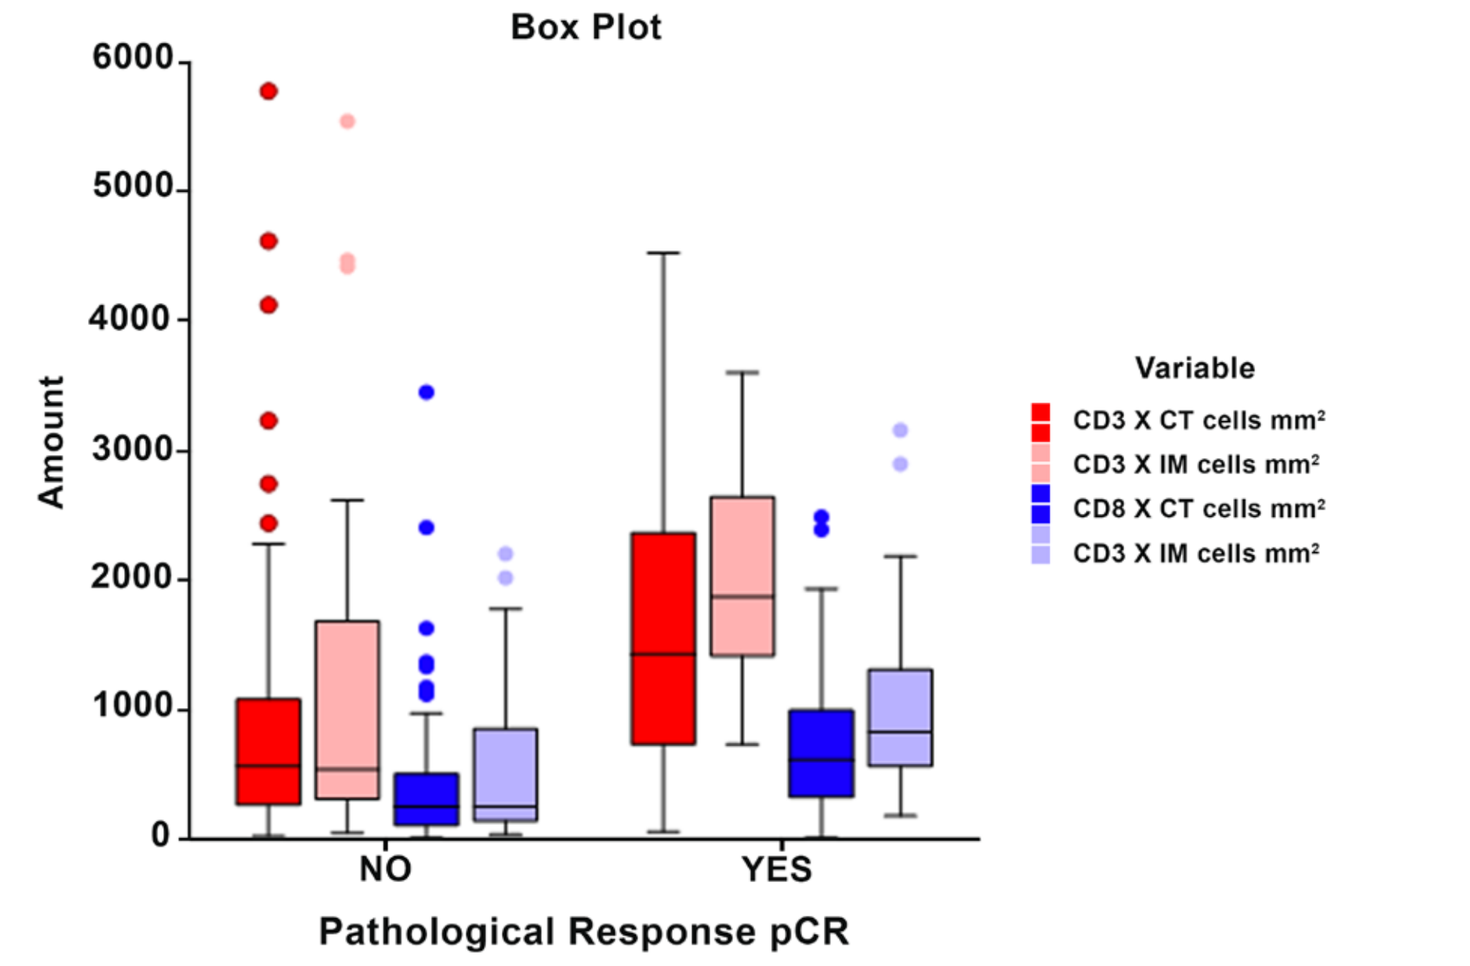
Box Plot
Amount
Variable
CD3 X CT cells mm²
CD3 X IM cells mm²
CD8 X CT cells mm²
CD3 X IM cells mm²
NO
YES
Pathological Response pCR

Table 3. Median cell density in patients with pCR vs non-pCR patients.

Median cell density in patients with pCR vs non-pCR patients

	Outcome	Median	CI (95,0%)	p-value
CD3 Centre of Tumour	No pCR	567,559	358,83 - 753.29	0,00329
	pCR	1432,01	1103,19 - 1900	
CD3 Invasive Margin	No pCR	540,828	431,97 - 1211.749	0,00043
	pCR	1877,745	1430,597 - 2418.445	
CD8 Centre of Tumour	No pCR	246,0505	154,086 - 307.483	0,01991
	pCR	614,485	450,177 - 749.512	
CD8 Invasive Margin	No pCR	255,148	175,811 - 425.343	0,00119
	pCR	827,267	643,216 - 1189.143	

Logistic regression analysis

Table 4. Logic regression analysis.

Coefficient Significance Tests

Independent	Regression Coefficient	Standard Error	Wald Z-value	Wald Prob	Odds Ratio
Ki-67 (Continuous)	5,84051	1,83561	3,182	0,00146	343,95612
Biological Type - Luminal	-2,79292	1,17165	-2,384	0,01714	0,06124
Immunoscore Intermediate	-1,80059	0,77698	-2,317	0,02048	0,1652
Immunoscore Low	-1,99918	0,98812	-2,023	0,04305	0,13545
Tumour 2-5cm	2,17458	1,09489	1,986	0,04702	8,79853
Biological Type - TNBC	-3,2585	1,66519	-1,957	0,05037	0,03845
Stage 2B	-2,58973	1,5177	-1,706	0,08794	0,07504
Stage 2A	-2,01162	1,25775	-1,599	0,10974	0,13377
Intercept	2,63261	1,67336	1,573	0,11566	13,91008
Stage 3	-2,83108	1,84867	-1,531	0,12567	0,05895
ER Positive	-1,63232	1,72928	-0,944	0,34521	0,19548
Tumour > 5cm	-1,37975	1,78558	-0,773	0,43969	0,25164
PR Positive	-0,85124	1,1142	-0,764	0,44487	0,42688

Conclusions

- Ki-67, Biological type, Immunoscore® and tumour size are independent prognostic factors of pCR in patients with early breast cancer undergoing neoadjuvant chemotherapy.

San Antonio Breast Cancer Symposium®; December 10 - 14, 2019 ; San Antonio, Texas, USA
This presentation is the intellectual property of the author/presenter. Contact them at brapoport@rosebankoncology.co.za for permission to reprint and/or distribute.

Corresponding author: brapoport@rosebankoncology.co.za